Comparison of the Activity of Myeloperoxidase Enzyme System to Antibiotics for Irrigation of Implant Material

Orthopaedic Research Society
50th Annual Meeting
March 7-10, 2004
San Francisco, CA
Poster# 1061

+*Robins, A; **Woodhead, S;***Denys, G;***Abril-Horpel, O;***Becquerelle, S;***Haag, W;***Valvani, S
+University of Washington, Seattle, WA **Ricerca Biosciences, Concord, OH ***Exoxemis, Inc., Little Rock, AR

Contact Information
www.exoxemis.com

Objectives

A newly formulated enzyme system containing myeloperoxidase (MPO) has been developed (ExOxEmis, Inc., Little Rock, AR) which has been shown to be rapidly bactericidal against an extensive range of microorganisms including biofilm producing strains of *Staphylococcus epidermidis* and *Pseudomonas aeruginosa*[1,2]. The objectives of this study were to (1) evaluate the potential efficacy of the *myeloperoxidase enzyme system* as an irrigation solution in the presence of implant material (polished stainless steel), and (2) compare its activity to traditional antibiotic containing solutions for the reduction or elimination of residual bacteria.

Methods

Organism. *Staphylococcus aureus* ATCC 29213 was used in this study. The organism was maintained at -70°C and subcultured on BBL trypticase soy agar plates (Becton Dickinson Microbiology Systems, Cockeysville, MD).

Antimicrobial agents. Myeloperoxidase containing formulation was prepared in our laboratory. Gentamicin (American Pharmaceutical Partners, Schaumberg, IL) and vancomycin (Abbott laboratories, North Chicago, IL) were obtained internally.

MIC determination. The minimal inhibitory concentration (MIC) of gentamicin (0.25 µg/ml) and vancomycin (0.5 µg/ml) was determined by the standard NCCLS[3] broth microdilution method.

Test design. The bacteriostatic or bactericidal effects of the *myeloperoxidase enzyme system* and comparator antibiotics were assessed by a time-kill assay. All time-kill assays were performed in parallel in scintillation vials with and without the addition of polished stainless steel (SS) coupons (9.53 mm dia. x 4.5 mm thick – 316L). A common bacterial suspension for each experiment was prepared from late log to early stationary phase growth to ~10^9 cfu/ml. The inoculum was then pelleted and resuspended in buffer.

•***Time-kill test for MPO***. A 2.0 ml volume of myeloperoxidase formulation with organism suspension adjusted to a final target concentration of ~10^6 cfu/ml was tested. The final myeloperoxidase concentrations in the formulation tested included 3, 6, 12, and 25 µg/ml. The vials were incubated at 37°C and the enzyme reactions stopped by the addition of catalase. Samples were then removed for viable counts at 30 sec, 1, 3, 5, 15, and 30 min. A control culture with no myeloperoxidase formulation was incubated for 30 min at 37°C and quantitative cultures performed.

•***Time-kill test for antibiotics***. A 6.0 ml volume of appropriate Mueller-Hinton broth (MHB), antibiotic, and organism suspension adjusted to a final target concentration of 10^6 cfu/ml was tested. The final antibiotic concentrations were 1x, 2x, 4x, and 8x the MIC. The vials were incubated at 37°C, and samples were removed for viable counts at 30 min, 4, 12, and 24 hrs. A starting growth control containing MHB, inoculum and antibiotic was included at 30 min and quantitative cultures were performed.

•***Quantitative cultures.*** At each appropriate time point, samples were collected and serially diluted in saline. A 100µl volume of each dilution was applied to trypticase soy agar and spread over the surface for quantitative culture. Following overnight incubation at 35°C, the colonies were counted and the viable count was calculated.

Results

•***Data analysis***. The results of the time-kill studies are summarized in Figure 1. The data is presented as $\log_{10}$ survivors after exposure to the *myeloperoxidase enzyme system* and antibiotics vs. time, with 10^0 representing 1 survivor. The breakpoint scale of no survivors represent the results obtained when the entire contents of the treatment vials were cultured.

•***MPO enzyme system.*** The *myeloperoxidase enzyme system* demonstrated rapid activity within 1.0 minute and was bactericidal within 15 minutes of treatment at the lowest concentration tested (3.0 µg/ml myeloperoxidase). At a MPO concentration of 25 µg/ml, complete kill was achieved within 3 min. Similar results were obtained in the presence of SS coupons in the reaction mixture indicating no interference with the performance of the enzyme formulation.

•***Antibiotics***. Gentamicin, a time-dependent antibiotic, demonstrated bacteriostatic activity (<3.0 log reduction) even at the highest concentrations tested. Vancomycin, a concentration-dependent antibiotic, demonstrated bactericidal activity ($\geq$3.0 log reduction) in the initial inoculum by 12 hours at 1.0-4.0 µg/ml. At 24 hours, a loss of activity was observed for both antibiotics due to re-growth of the initial inoculum.

Figure 1.
Comparison of the bactericidal activity of *myeloperoxidase enzyme system* versus bacteriostatic activity of gentamicin and vancomycin against *Staphylococcus aureus*

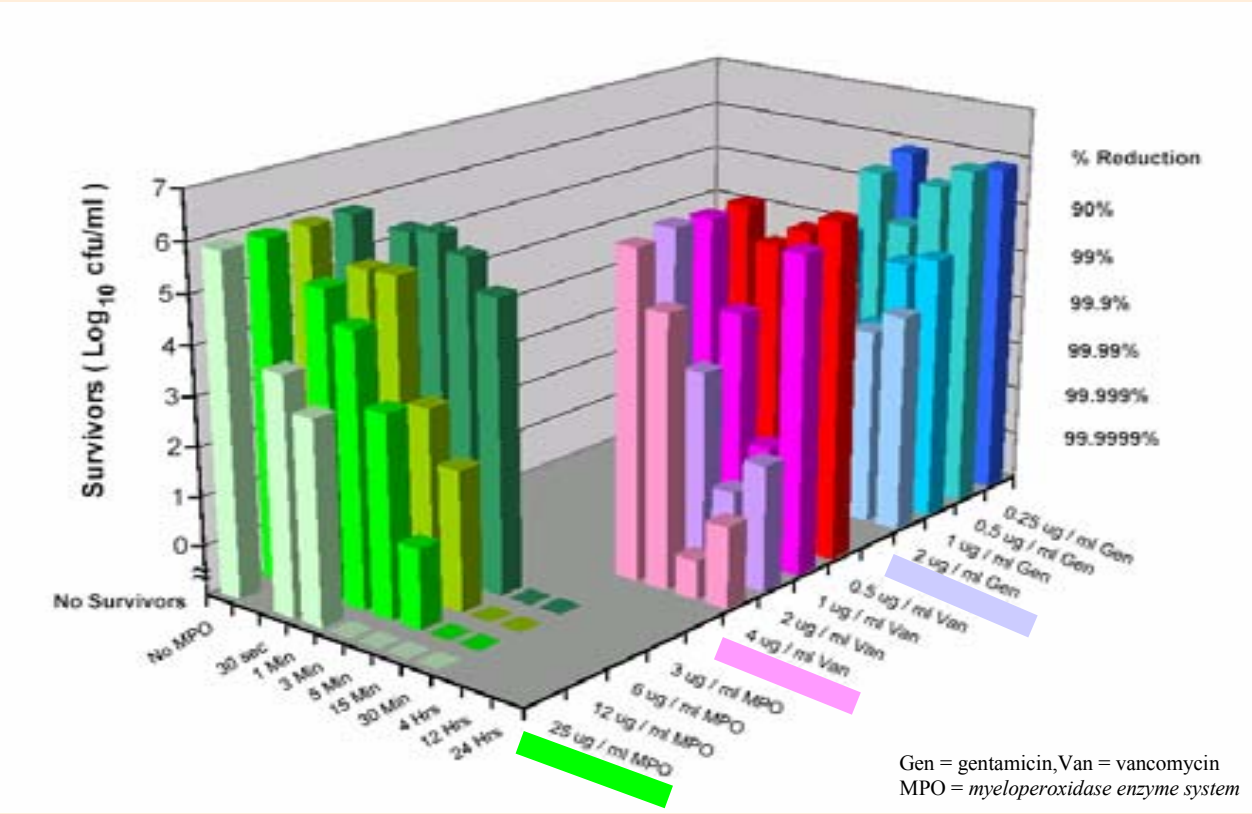
Survivors (Log10 cfu/ml)
7
6
5
4
3
2
1
0
No Survivors
No MPO
30 sec
1 Min
3 Min
5 Min
15 Min
30 Min
4 Hrs
12 Hrs
24 Hrs
25 ug / ml MPO
12 ug / ml MPO
6 ug / ml MPO
3 ug / ml MPO
4 ug / ml Van
2 ug / ml Van
1 ug / ml Van
0.5 ug / ml Van
2 ug / ml Gen
1 ug / ml Gen
0.5 ug / ml Gen
0.25 ug / ml Gen
% Reduction
90%
99%
99.9%
99.99%
99.999%
99.9999%
Gen = gentamicin,Van = vancomycin
MPO = *myeloperoxidase enzyme system*

Conclusions

• The *myeloperoxidase enzyme system* demonstrated both rapid (less than or equal to 15 minutes) and complete kill of residual *S. aureus*, even in the presence of SS implant material.

• The antibiotic solutions failed to demonstrate or sustain bactericidal activity as shown by re-growth of the initial inoculum.

• The *in-vitro* efficacy of *myeloperoxidase enzyme system* demonstrated superiority over agents used in current clinical practice for reducing residual bacteria.

• Additional studies are needed to assess the efficacy of the *myeloperoxidase enzyme system* with other pathogens, implant materials and surfaces, and to determine if the enzyme formulation can prevent bacterial attachment to implant material and biofilm formation.

References

(1)Woodhead,S., Denys, G., Harrell, L., Hill, G., Valvani, S., Abril-Horpel, O., and Haag, W., 2003. Microbicidal activity of the myeloperoxidase enzyme system against clinically relevant bacterial and fungal isolates including multidrug-resistant strains. American Society for Microbiology, 43rd ICAAC, Chicago, IL, Abst. F-1458.
(2)Robins, A., and Woodhead, S. 2003. A new approach for the prevention and treatment of staphylococcal musculoskeletal infection. Orthopaedic Research Society, 49th Annual Meeting, New Orleans, LA, Abst. 1062.
(3)National Committee for Clinical Laboratory Standards. 2003. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. Approved standard M7, 6th ed. National Committee for Clinical laboratory Standards, Wayne, Pa.

50th Annual Meeting of the Orthopaedic Research Society
March 7-10, 2004
San Francisco, CA